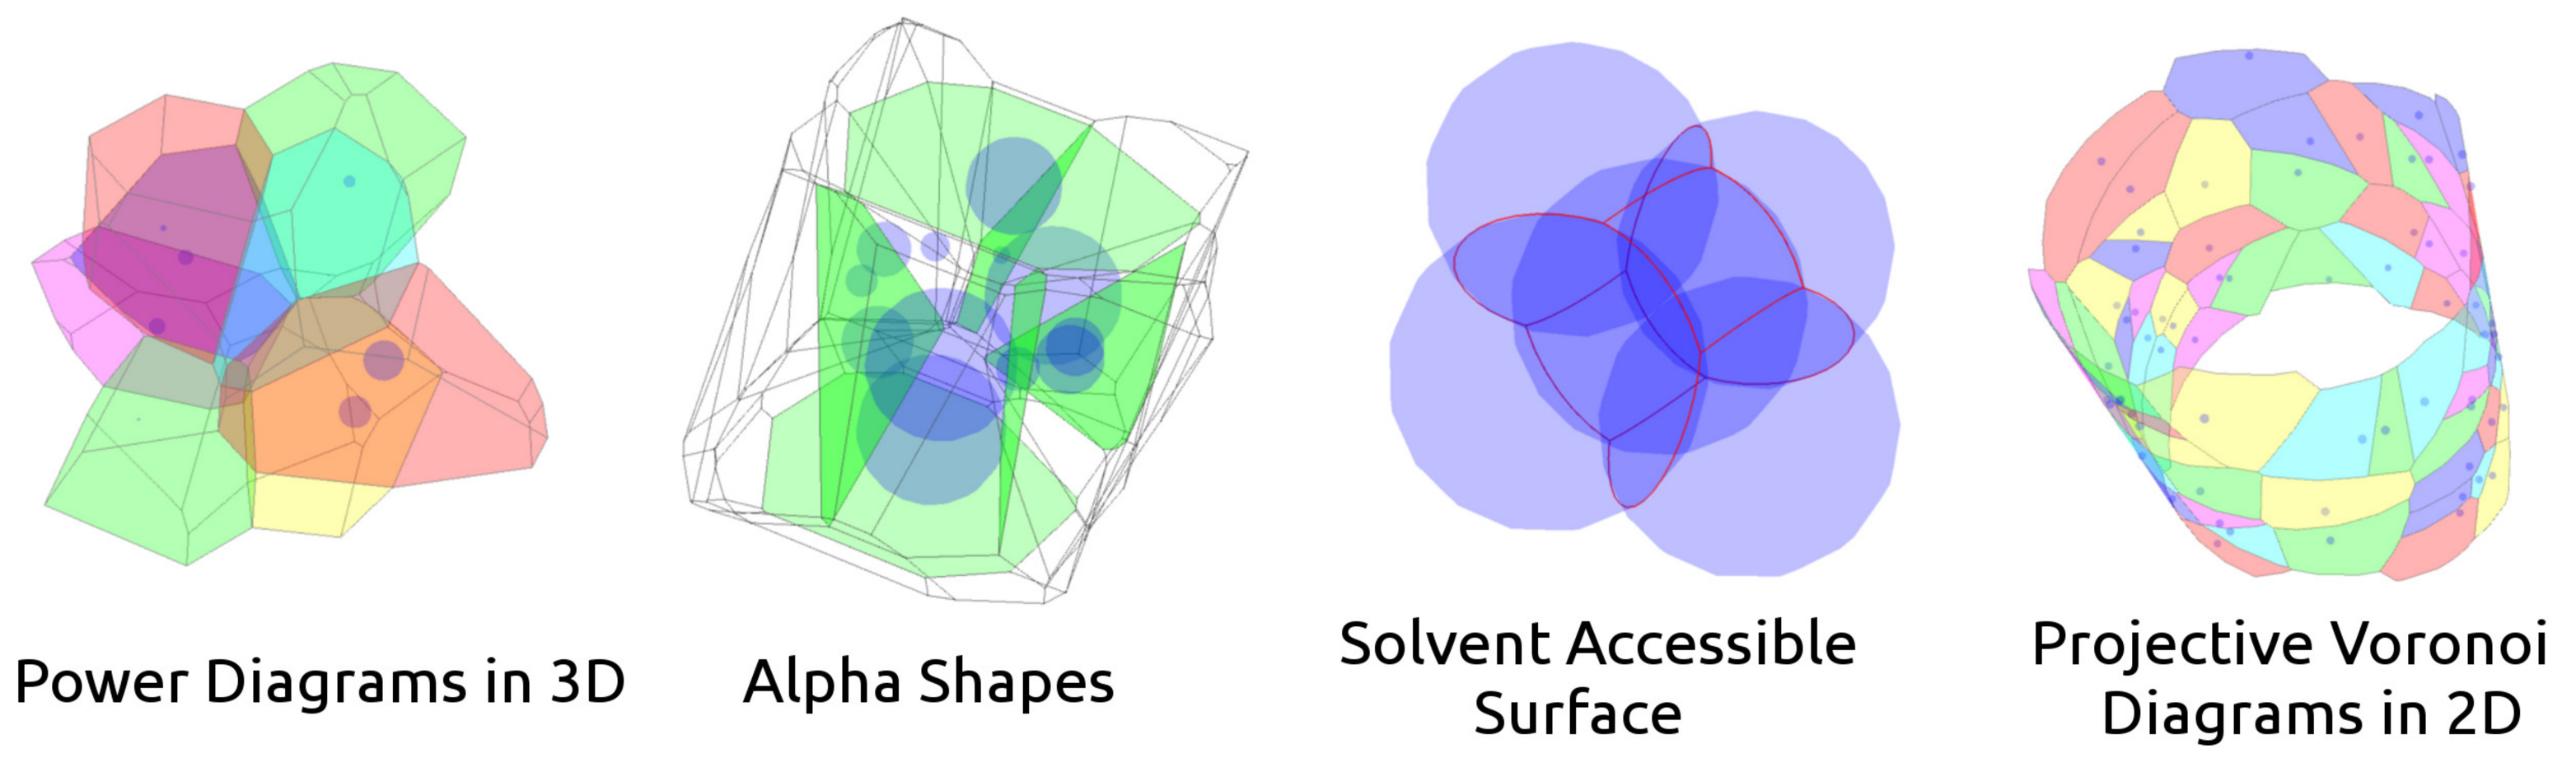

Power Diagrams in 3D
Alpha Shapes
Solvent Accessible Surface
Projective Voronoi Diagrams in 2D

DMG-α: computational geometry package for analysis of molecular dynamic simulations

Robert Szczelina[1,2], Krzysztof Murzyn[2]

1. Jagiellonian University, Faculty of Math. and Comp. Science, Department of Comp. Science,
2. Jagiellonian University, Malopolska Center of Biotechnology, robert.szczelina@uj.edu.pl,
3. Jagiellonian University, Faculty of Biochemistry, Biophysics, and Biotechnology, Department of Computational Biophysics and Bioinformatics

Introduction

The DMG-α library grants researchers in the field of computational biology, chemistry, and biophysics access to an open-sourced, easy to use and intuitive software for performing fine-grained geometric analysis of molecular systems. The library is capable of computing power diagrams (weighted Voronoi diagrams) in three dimensions with 3D periodic boundary conditions, computing approximate projective 2D Voronoi diagrams on arbitrarily defined surfaces, performing shape properties recognition using α-shape theory, and can do exact Solvent Accessible Surface Area (SASA) computation. A rich Python interface (`pydmga`) is provided as a convenient way to manipulate the DMG-α routines (implemented in C++ for performance). We illustrate possible applications of the DMG-α to determine non-trivial geometric properties of two *Escherichia coli*-specific lipids as emerging from molecular dynamics simulations of relevant model bilayers.
DMG-α along with all scripts are available for download under terms of BSD license to anyone interested from the project webpage: `http://dmgalpha.scirsc.org`.

DMG-α Library

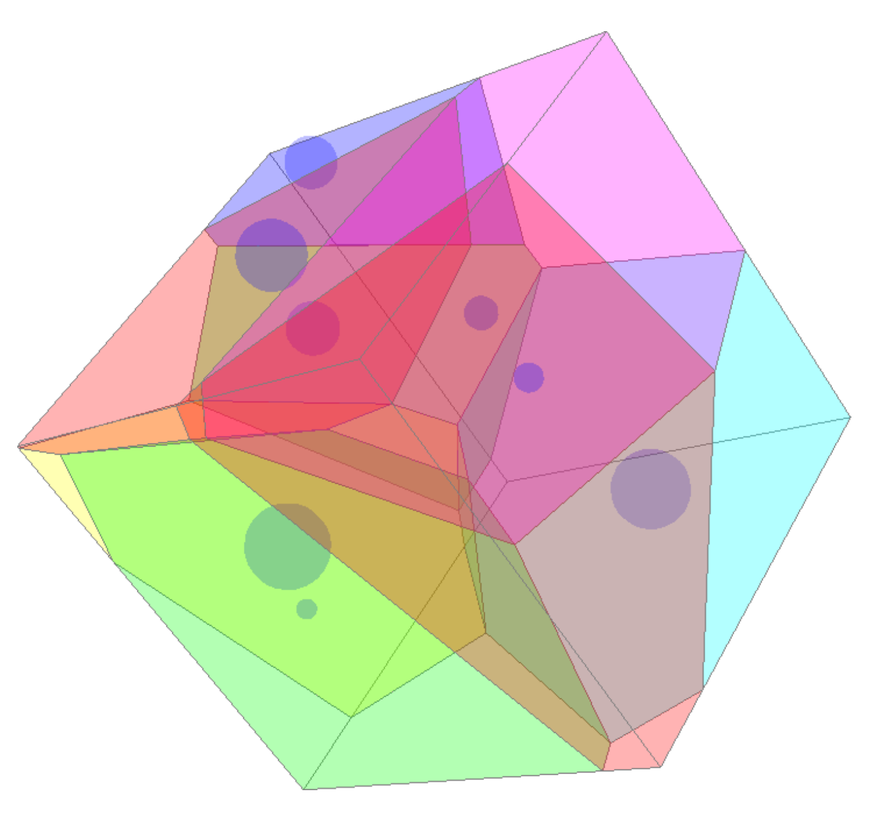

A Power Diagram

The Power Diagram (PD) of a set of n balls in 3D assigns to each of them a convex polyhedron (*a power cell*) that contains all points in the space that are closer to this ball than to any other (w.r.t. some pseudo-distance function).

Two cells sharing a common face are called *adjacent* and this relation allows to rigorously investigate inner topological structure of the (union of) set of balls. PD can be used to infer data structures and quantities such as α-shapes (to identify pockets and tunnels), Solvent Accessible Surface Area (SASA) and the volume of the union of balls.

PD may be hard to compute in higher dimensions - optimal algorithms for a set of n balls in d-dimensional space needs time proportional to $n^{\frac{d}{2}}$ in the worst case. However, in the context of large, uniformly distributed sets inside a compact subset of R^3, expected running time is proportional to n. Therefore it is feasible to compute PD for sets originated from Molecular Dynamic (MD) simulations, especially for solvated models of biomolecules.

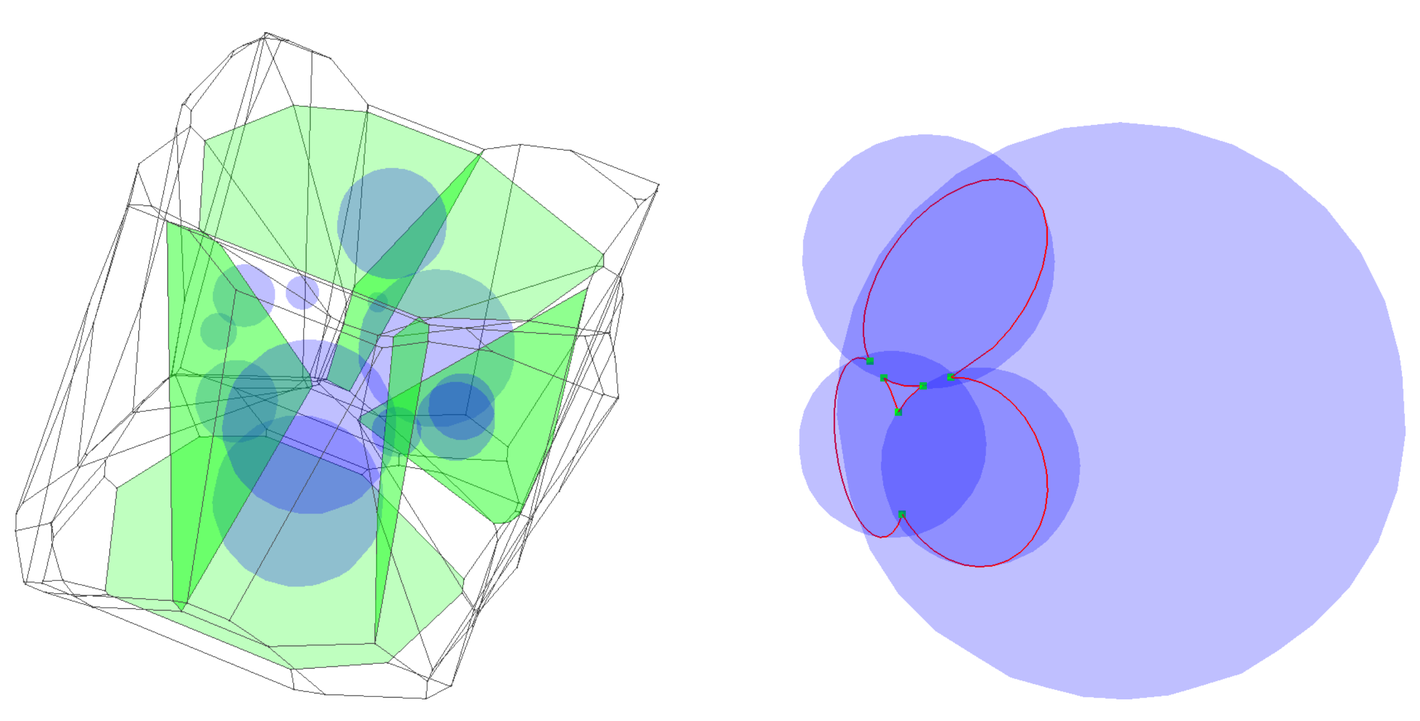

alpha shape (left) and SA Surface (right).

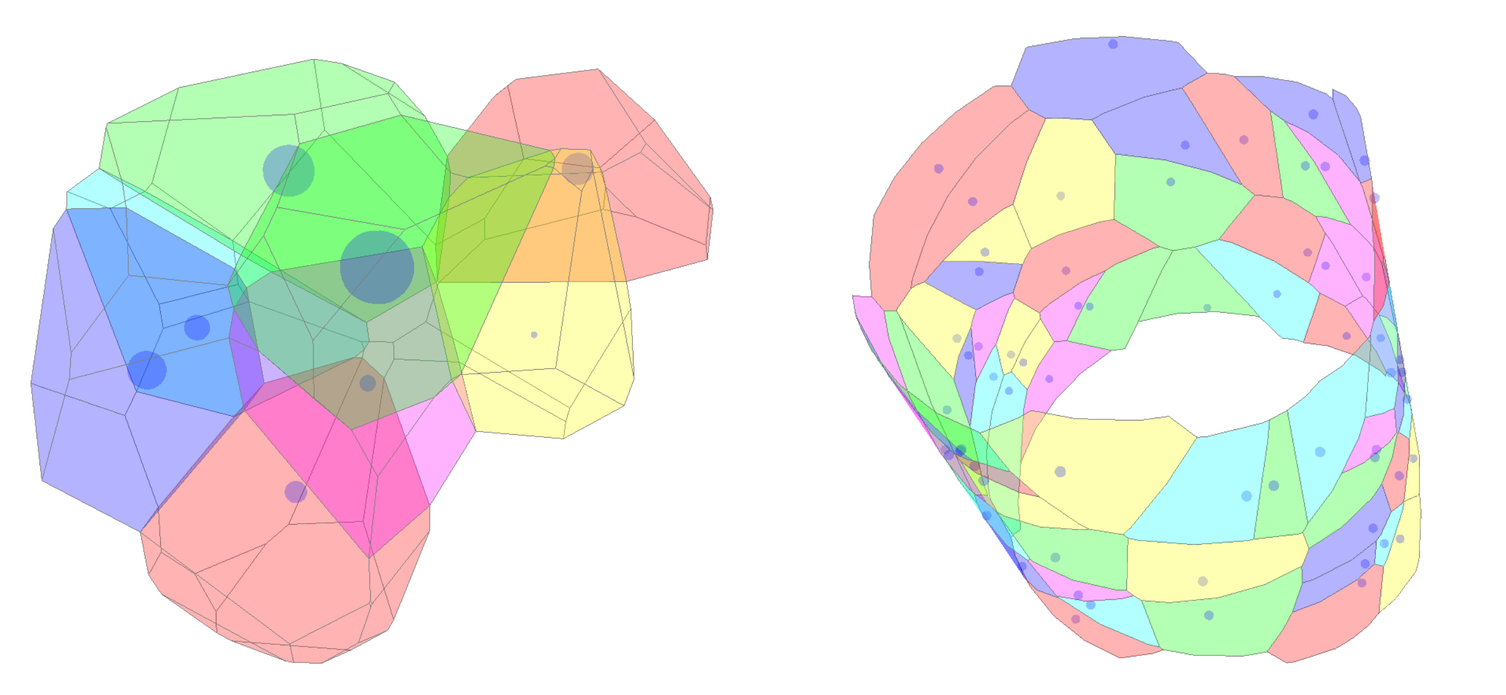

PBC in PD (left) and 2D PB on a cylinder (right).

In the case of molecular systems periodic boundary conditions (PBC) are of common use. It is essential to efficiently compute PD constrained to a simulation box and to develop all algorithms (PD, SASA, α-shapes) that take PBC into consideration. Also, it is useful to be able to compute 2D PD of the projection onto arbitrary surface.

DMG-α implements all mentioned algorithms and wrap them in user-friendly, scriptable Python interface. The performance tests of the library shows good linear dependence on the size of the input as well as on the number of parallel computational cores.

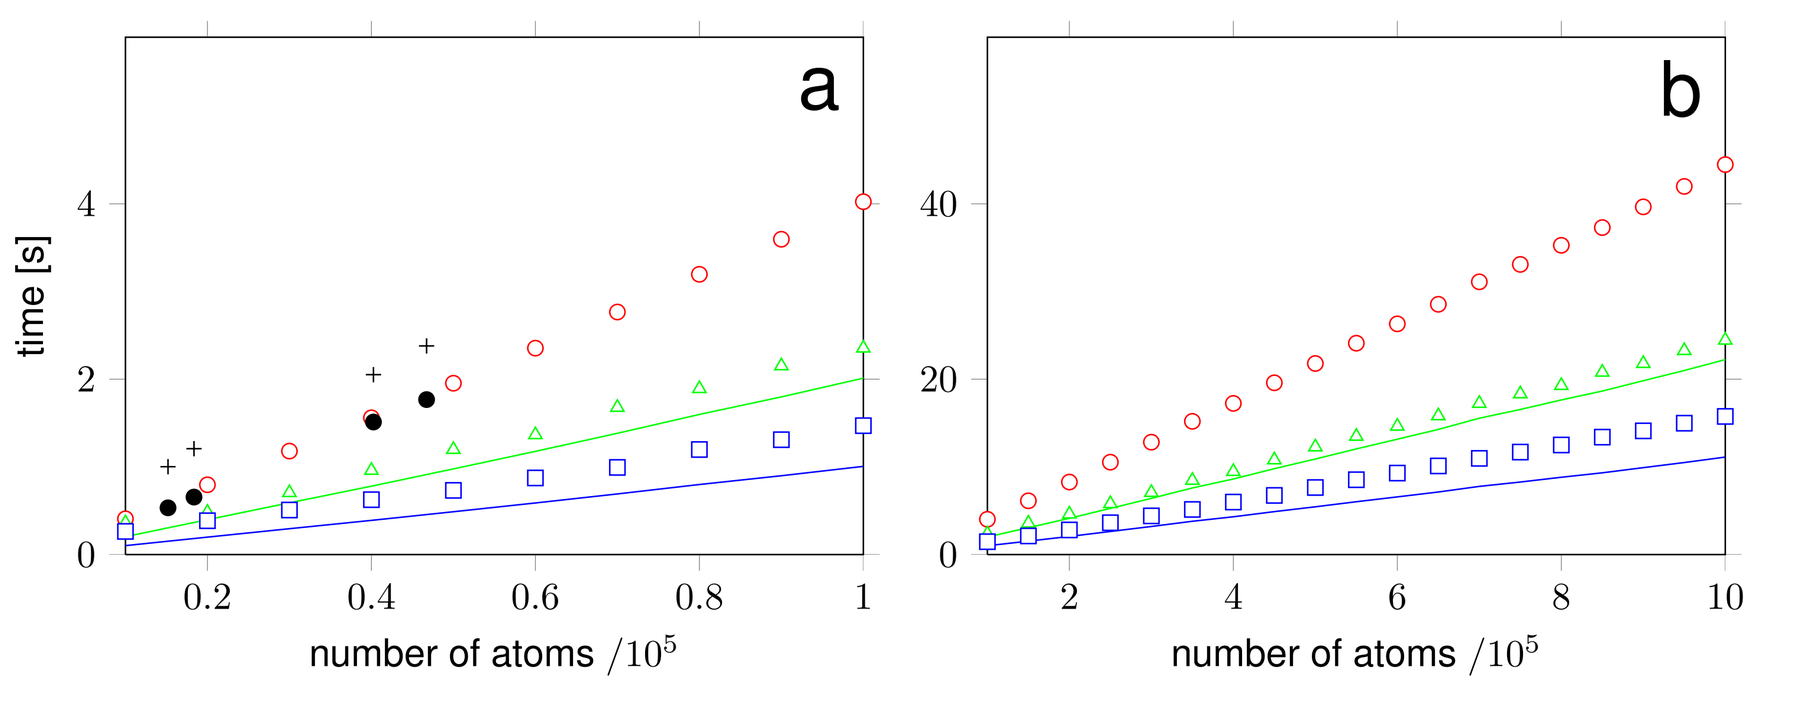

a
b
time [s]
number of atoms /10^5
number of atoms /10^5

Performance tests of DMG-α lib. Symbols represent speed for a single test while lines present best possible speed-up for a given number of cores (resp. 2 and 4).

References

[1] Robert Szczelina, Krzysztof Murzyn, *DMG-α – a computational geometry library for multi-molecular systems*, submitted to Journal of Chemical Information and Modeling (2014)

Robert Szczelina and Krzysztof Murzyn acknowledge financial support from Polish National Science Centre under grants no. 2011/01/N/ST6/07173 and 2011/01/B/NZ1/00081, respectively.

This poster was typeset using *baposter* class `http://www.brian-amberg.de/uni/poster/`

Application: E.Coli model membranes

INTRODUCTION: We determined several geometric properties of two fully hydrated lipid bilayers in the liquid crystalline phase from the outer bacterial membrane of *E. coli*: Lipid-A (ECLA) and deep-rough lipopolysaccharide (DREC).
METHODS: OPLS/AA (lipids) forcefield parameters and TIP3P (water) parameters were used. The charge of ECLA ($-2e$) and DREC ($-4e$) molecules was counterbalanced by sodium cations. The total number of atoms model systems was 40248 (ECLA) and 46728 (DREC), respectively. Submicrosecond MD simulations were carried out in Gromacs v4.5. For analysis, we used 11 consecutive frames, sampled every one nanosecond, from fully developed MD trajectories.
RESULTS: V_m of ECLA molecules is significantly smaller than in DREC molecules, which is the expected result in view of the apparent differences in their headgroups. The average values of V_m for ECLA and DREC are, respectively, 2.86 and 3.27 nm^3. The similar pattern is seen for SASA. The DREC molecule with the larger headgroup has larger SASA. The average values of SASA for ECLA and DREC are, respectively, 4.15 and 6.82 nm^2. Interestingly, distribution of SA for ECLA and DREC bilayers are very similar with the average values, respectively, 1.50 and 1.55 nm^2. This clearly indicates that the main difference is not in SA but in the width of lipid/water interphase. Other differences in the structure of lipid/water interphase of ECLA and DREC are very well seen in the number of water and ions interacting directly with membrane. ECLA and DREC molecules make contacts on average with 2.35 and 6.43 ions, respectively. Distribution of water molecules forming the first hydration layer is also distinctly different with the average water molecules per lipid equal to, 47.3 and 68.8, respectively.

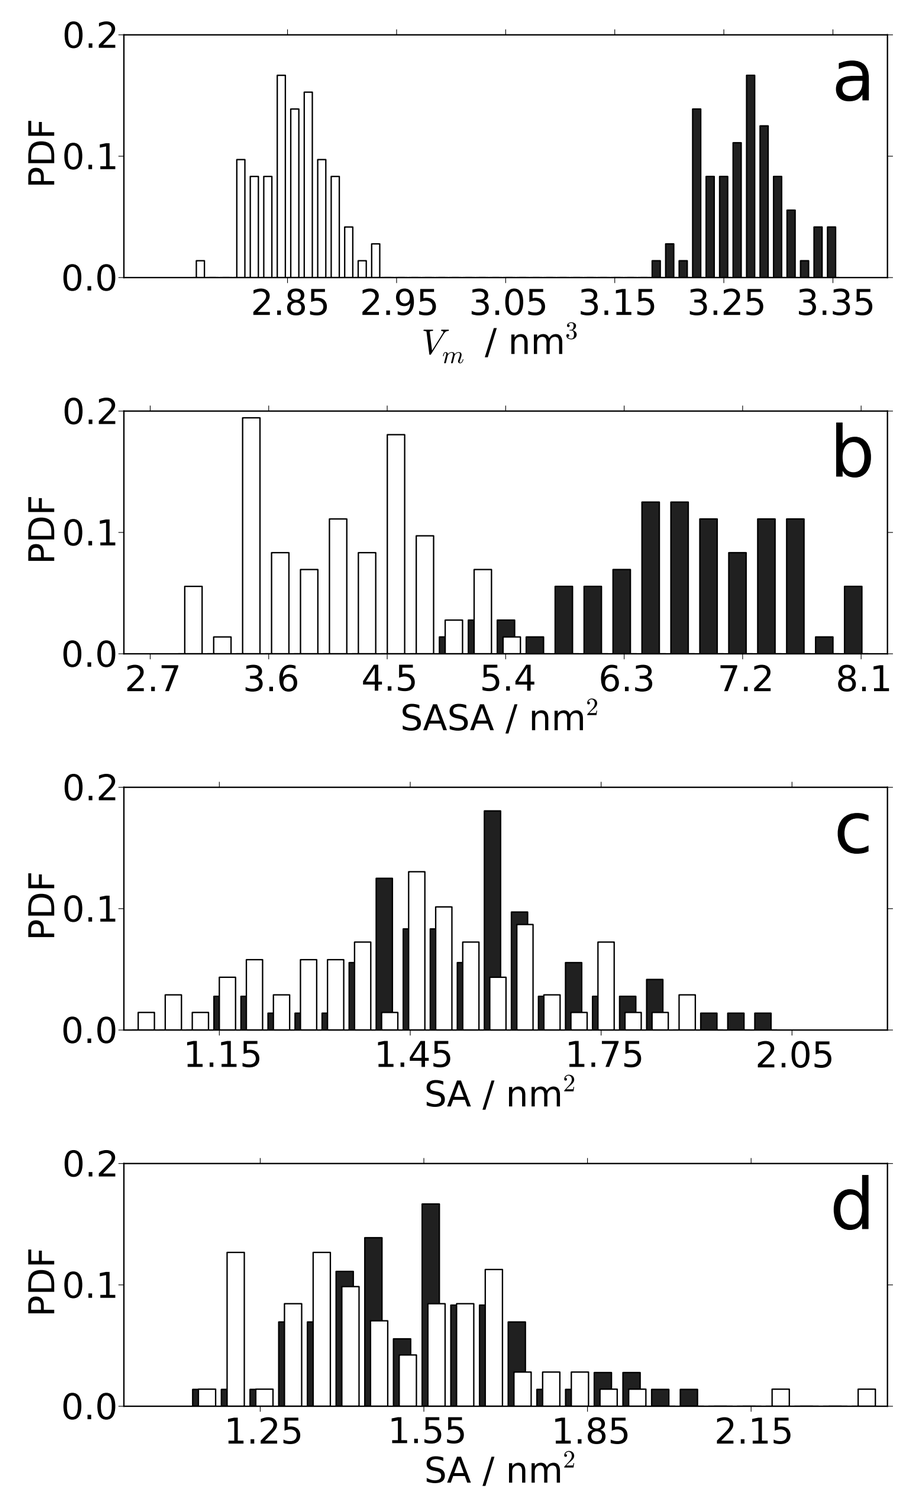

a
b
c
d
PDF
V_m / nm^3
SASA / nm^2
SA / nm^2
SA / nm^2

avg. volume (a), SASA (b), SA - two methods (c, d) per lipid molecule

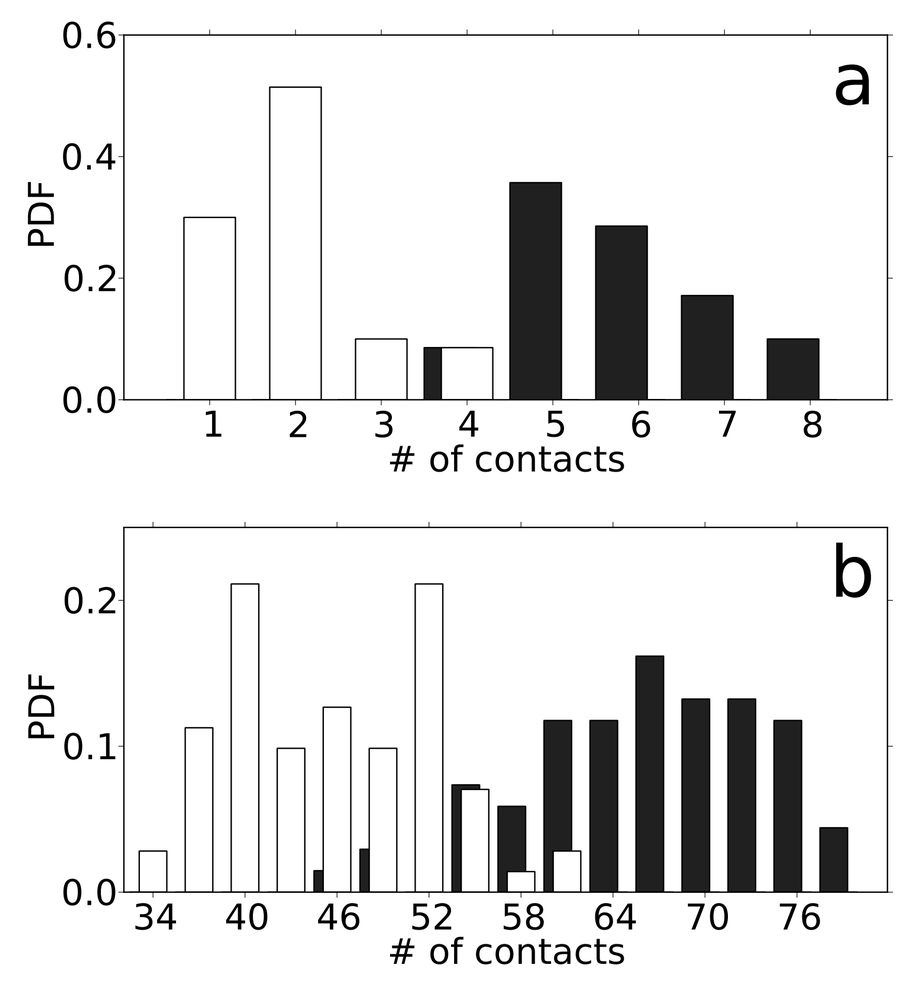

a
b
PDF
of contacts
of contacts

avg. # of ions (a) and H_2O (b) in contact with lipid molecule

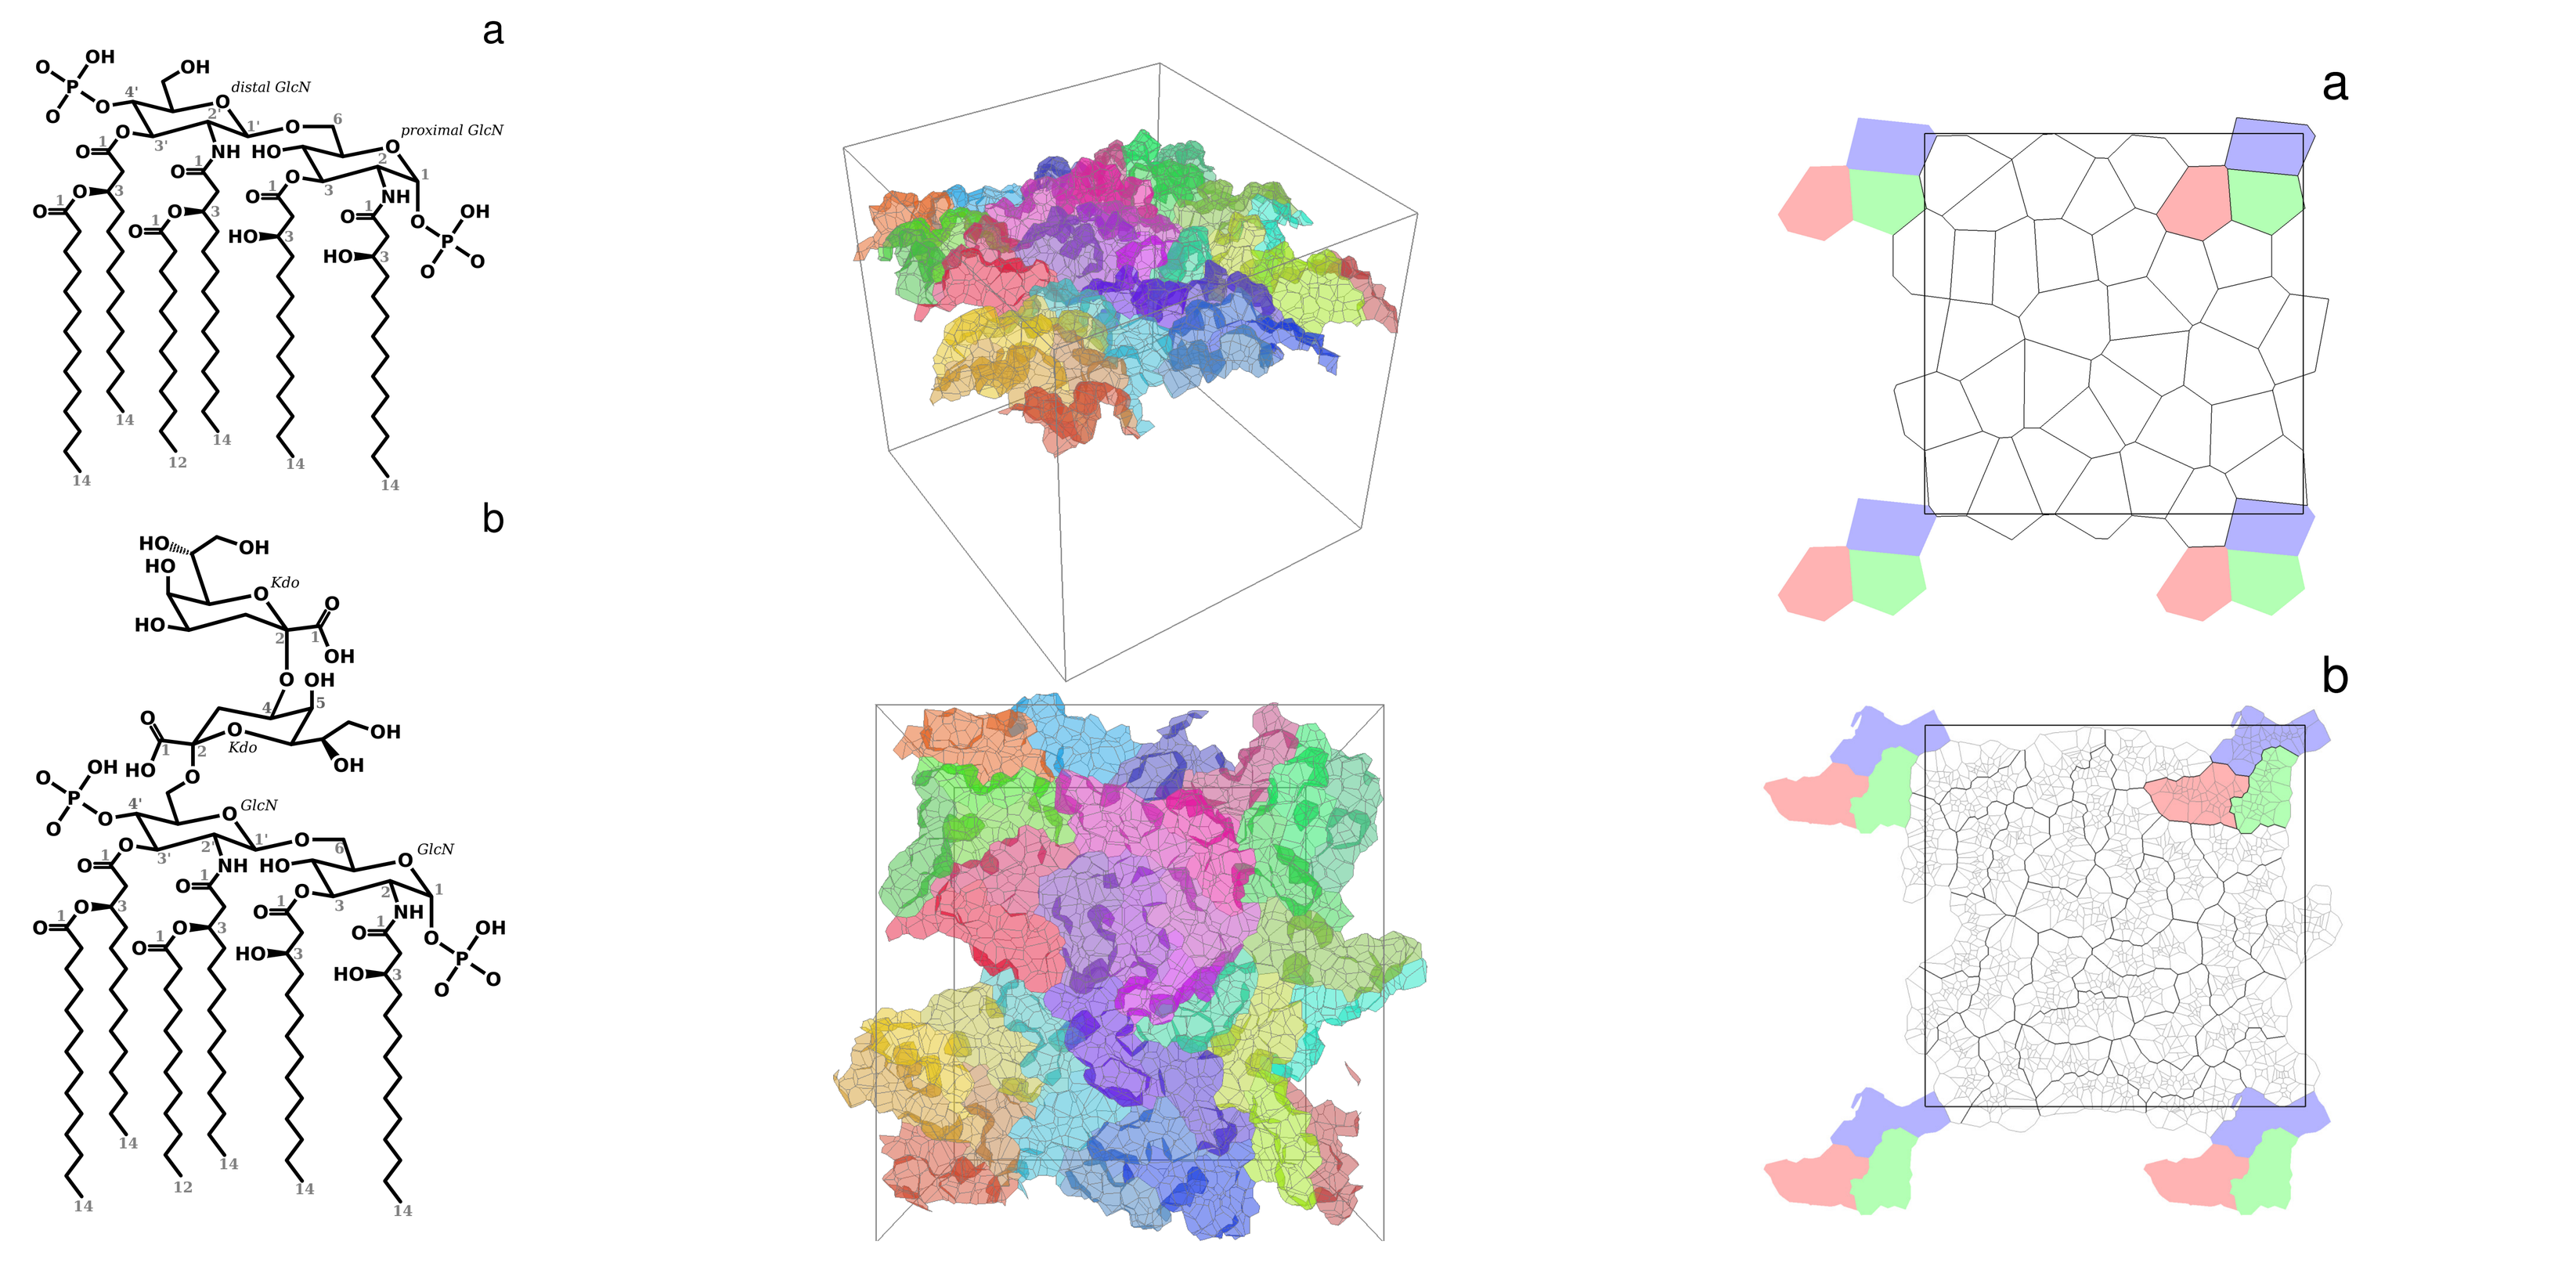

a
b
a
b

Left: The chemical structure of ECLA and (b) DREC lipids investigated in [1]. Center: reconstruction of the Surface of ECLA using DMG-α library. Right: two possible ways to compute SA (a) center of a headgroup, (b) all atoms of a headgroup.

Sample code: analysis of DREC trajectory

```
hao = True; end_lipid_at = "UDC"; start_lipid_at = "KDO"
in_file = file("drec-1_590-600ns.pdb", "r")
out_file = file("drec-1_590-600ns.dat", "w")
try:
  frame = 0
  out_file.write("frame␣lipid␣volume␣area\n")
  while True:
    particles = []
    while True:
      item = pdb.PDBRecord(pdb.readnext(in_file))
      if item.type() == "CRYST1":
        geometry = OrthogonalGeometry(item["a"], item["b"], item["c"], True, True, True)
        container = Container(geometry)
      if item.type() == "ATOM":
        if (item["atom"][0] != 'H' and item["atom"][1] != 'H') or not hao:
          container.add(item.as_particle())
          particles.append(item)
      if item.type() == "ENDMDL" or item.type() == "TER":
        break
    diagram = Diagram(container)
    molecule_no = 0
    residue = end_lipid_at
    for i, cell in enumerate(diagram):
      if particles[i]["residue"] == "SOL" or particles[i]["residue"] == "ION":
        break
      if residue == end_lipid_at and particles[i]["residue"] == start_lipid_at:
        if molecule_no: out_file.write("{0}␣{1}␣{2}␣{3}\n".format(frame, molecule_no, volume, area))
        molecule_no +=1
        area = 0.0
        volume = 0.0
      residue = particles[i]["residue"]
      volume += cell.volume()
      for side in cell.sides:
        neighbour = side.neighbour
        if particles[neighbour]['residue'] == "SOL" or particles[neighbour]['residue'] == "ION":
          area += side.area()
    out_file.write("{0}␣{1}␣{2}␣{3}\n".format(frame, molecule_no, volume, area))
    frame += 1
except StopIteration:
  pass
```